Frequent Dosing and GPNMB Expression with CDX-011 (CR011-vcMMAE), an Antibody-Drug Conjugate (ADC), in Patients with Advanced Melanoma

O. Hamid[1], M. Sznol[2], A. C. Pavlick[3], H. M. Kluger[2], K. B. Kim[4], P. D. Boasberg[1], R. Simantov[5], T.A. Davis[5], E. Crowley[5], T. Hawthorne[5], J. Green[5], P. Hwu[4]

[1]The Angeles Clinic and Research Institute, Santa Monica, CA; [2]Yale Cancer Center, New Haven, CT; [3]New York University Medical Center, New York, NY; [4]University of Texas MD Anderson Cancer Center, Houston, TX; [5]Celldex Therapeutics, Inc., Needham, MA

BACKGROUND

CDX-011 (glembatumumab vedotin) is designed to be stable in the bloodstream, but to release MMAE upon internalization into GPNMB-expressing tumor cells, resulting in a targeted cell-killing effect.

- Linker-MMAE technology, as used in brentuximab vedotin (SGN-35), licensed from Seattle Genetics.

CDX-011: Antibody-Drug Conjugate

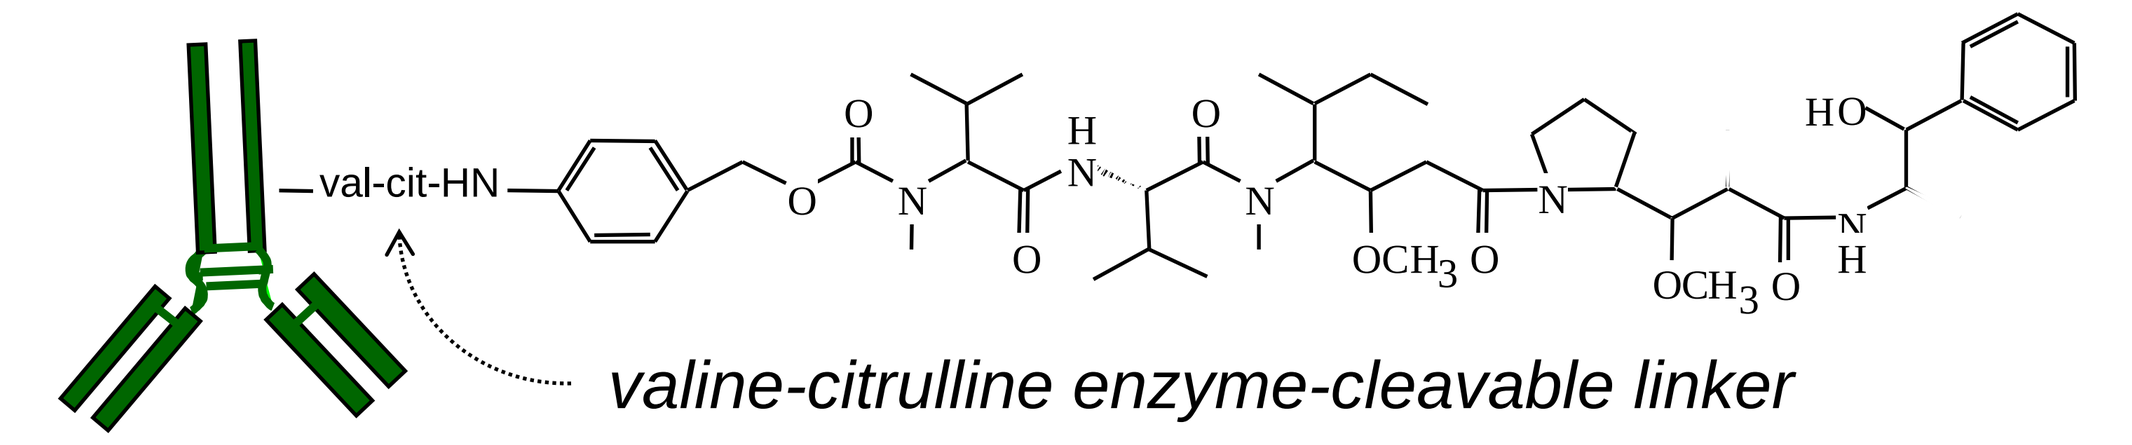
val-cit-HN
valine-citrulline enzyme-cleavable linker

CR011: fully-human IgG_2 targeting GPNMB

MMAE: dolastatin-like tubulin inhibitor

GPNMB identified by high throughput genomic screening; overexpressed in melanoma and other cancers

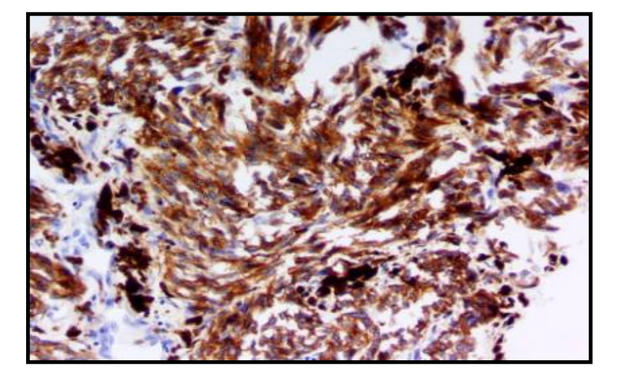

GPNMB expressed in 80% of human melanoma specimens

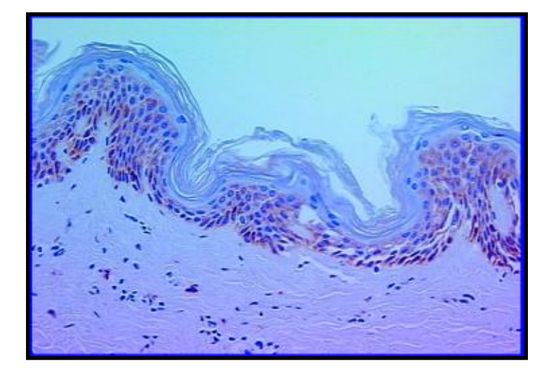

IHC shows some staining of pigmented epithelium in normal human skin

ERK Pathway Inhibitors and Imatinib Upregulate GPNMB and Sensitize Melanoma Cells Lines to CDX-011[1]

- ERK inhibitors and imatinib upregulate GPNMB expression on melanoma cell lines in vitro
- Effect of ERK inhibitors was dependent on cell lines harboring NRAS or BRAF mutations
- Pre-treatment with ERK pathway inhibitors augmented tumor growth inhibition by CDX-011 in vitro
- Potential for synergistic activity in the clinic

[1] Qian, et. al., Mol Oncology, 2:91:2008

In a Phase I/II study in patients with heavily pre-treated, advanced breast cancer (abstract #1095), CDX-011 has been shown to be active with observed objective responses. The current Phase I/II study evaluates the safety and efficacy of CDX-011 in metastatic melanoma.

GPNMB DETECTION

- Tumor tissue was analyzed for GPNMB expression at a centralized laboratory via IHC, using a biotinylated CR011 monoclonal antibody.
- Tumor tissue for 33 of 42 (79%) patients analyzed to date are positive for GPNMB expression.

STUDY DESIGN AND CONDUCT

Patient Population

- Unresectable Stage III or Stage IV melanoma
- Progressive disease at entry
- Karnofsky PS ≥70%
- ≤ 1 prior cytotoxic regimen
- Any number of cytokine, immune or vaccine therapies

Study Design

- Multicenter, Open-label, Phase I/II Trial
- CDX-011: 90 minute IV infusion
- Every three week (q3w) dosing schedule:
 - Sequential dose-escalation cohorts (n=3-6 patients)
 - Phase II expansion at MTD
 - ♦ Primary activity endpoint: Objective Response Rate (p_0=5%; p_1=20%; α=β=0.10)
- Subsequent evaluation of more frequent dosing schedules:
 - Once weekly for 2 of 3 weeks (q2/3w) and weekly (qw)
 - Sequential dose-escalation cohorts (n=3-6 patients)
 - Option to expand (n=15) at MTDs

Every Three Week (q3w) Dosing Schedule

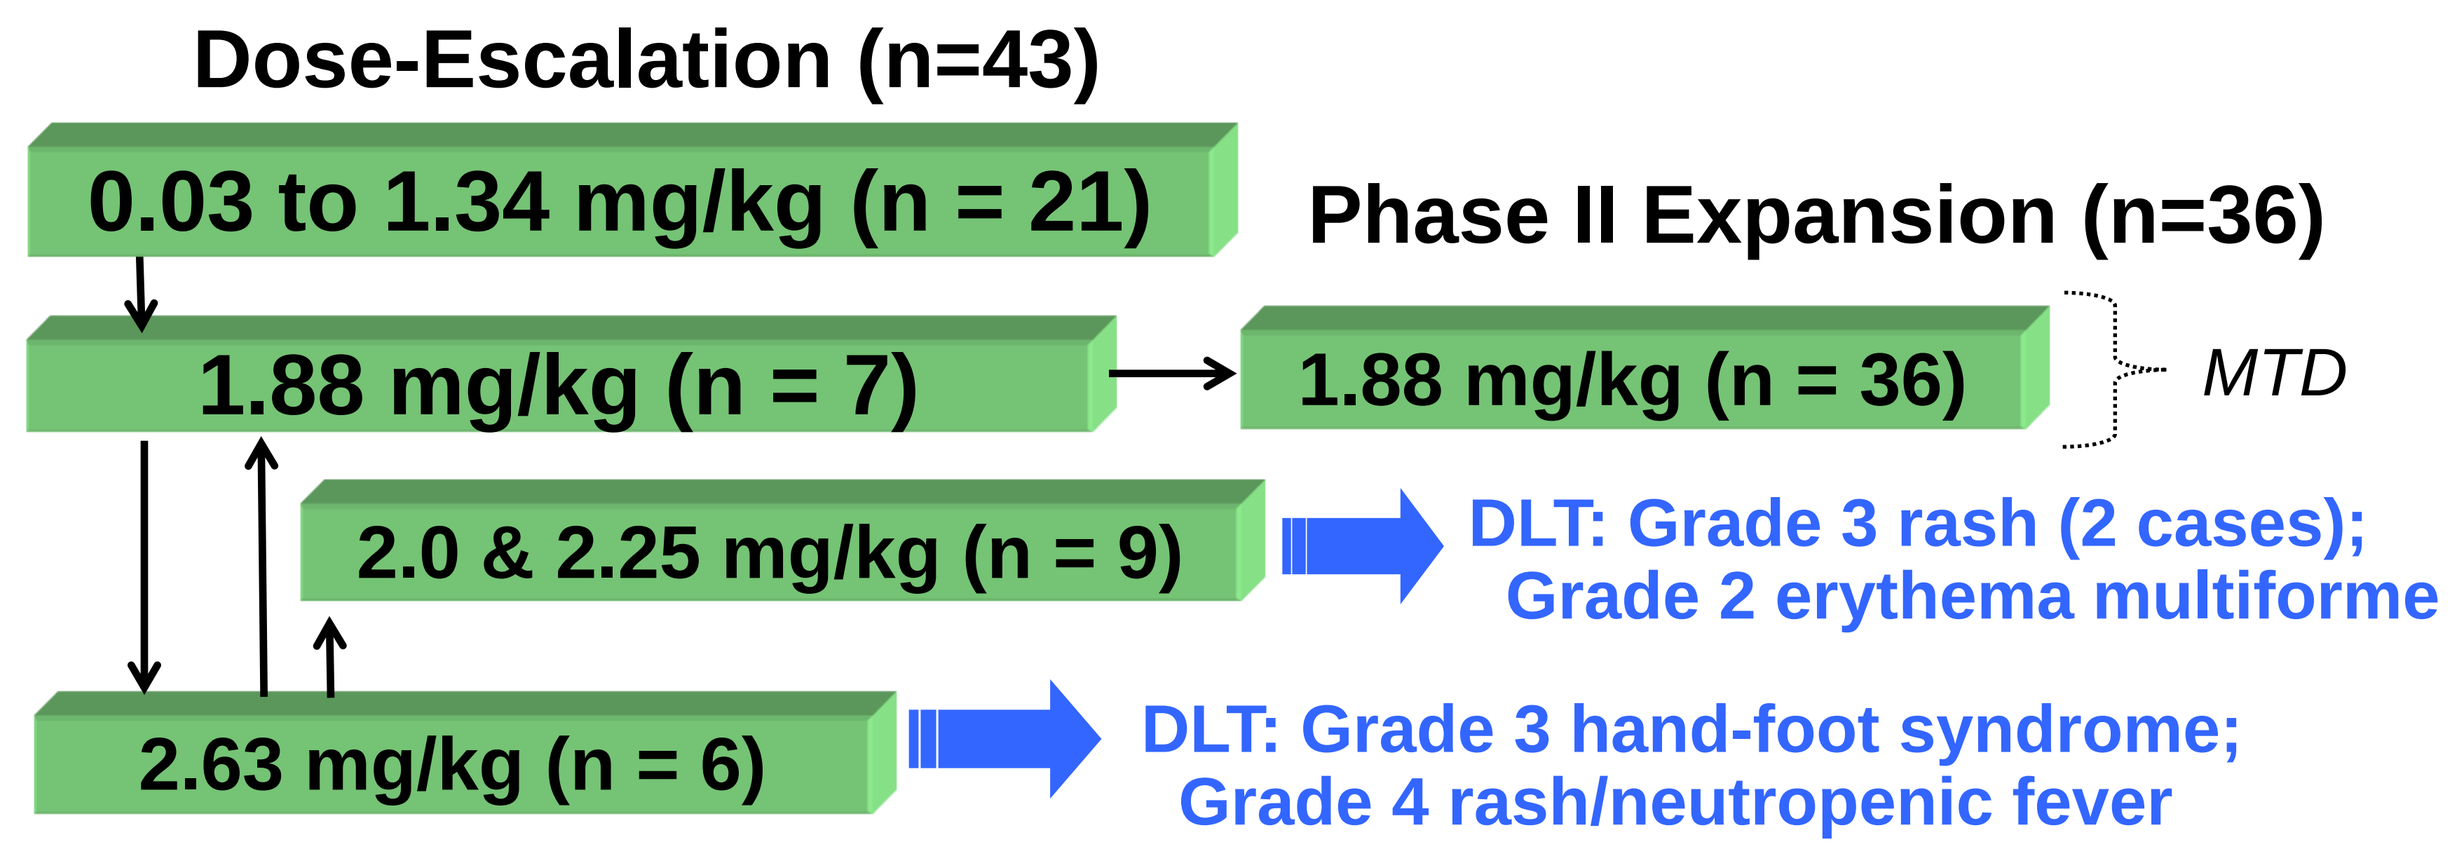
Dose-Escalation (n=43)
0.03 to 1.34 mg/kg (n = 21)
Phase II Expansion (n=36)
1.88 mg/kg (n = 7)
1.88 mg/kg (n = 36)
MTD
2.0 & 2.25 mg/kg (n = 9)
DLT: Grade 3 rash (2 cases); Grade 2 erythema multiforme
2.63 mg/kg (n = 6)
DLT: Grade 3 hand-foot syndrome; Grade 4 rash/neutropenic fever

Two of Three Week (q2/3w) Dosing Schedule

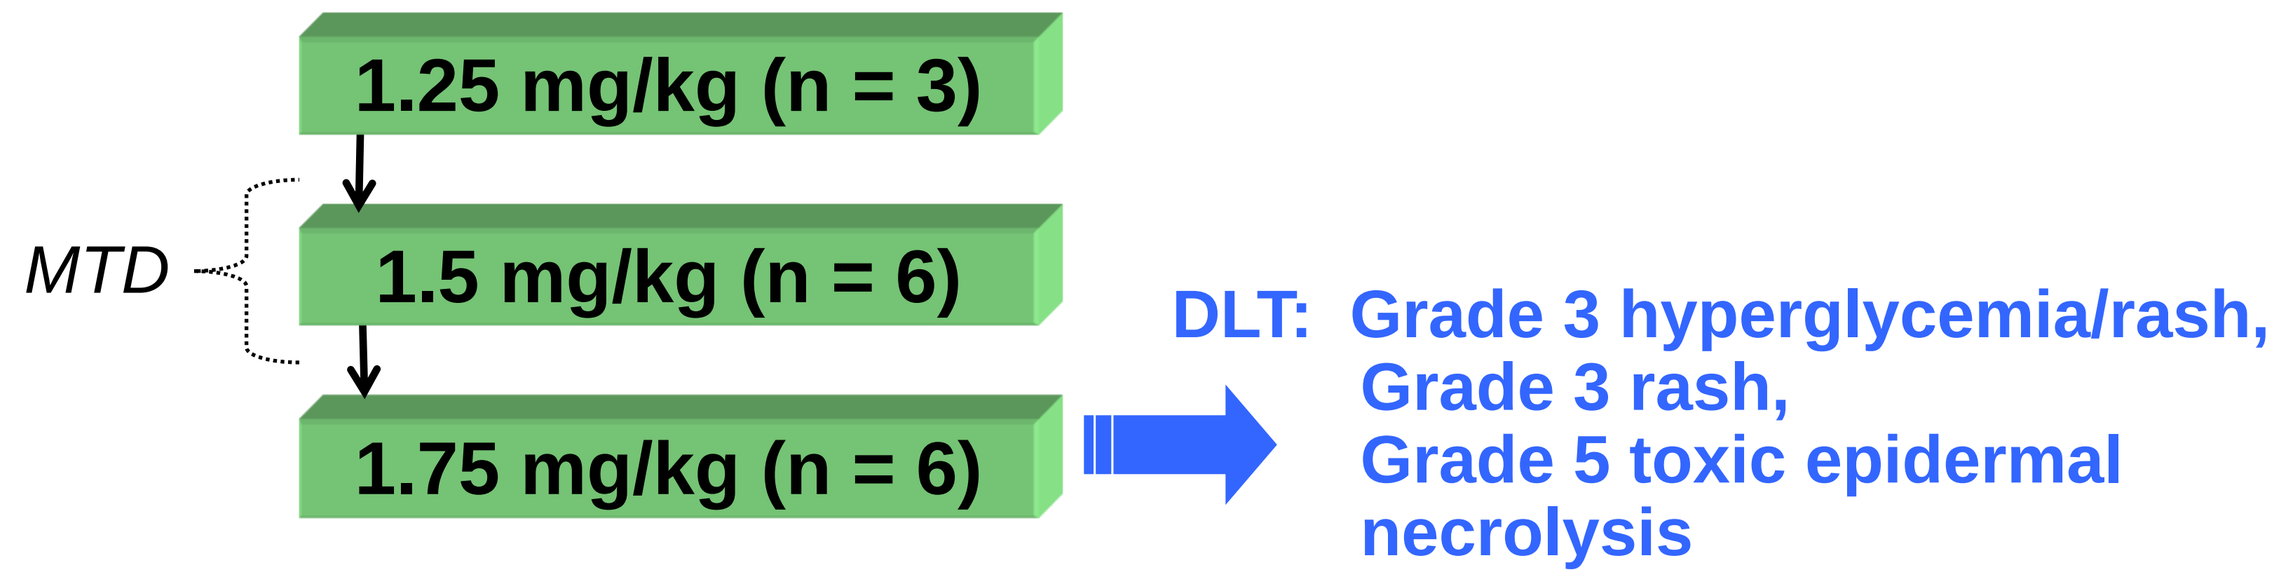
1.25 mg/kg (n = 3)
MTD
1.5 mg/kg (n = 6)
1.75 mg/kg (n = 6)
DLT: Grade 3 hyperglycemia/rash, Grade 3 rash, Grade 5 toxic epidermal necrolysis

Weekly (qw) Dosing Schedule

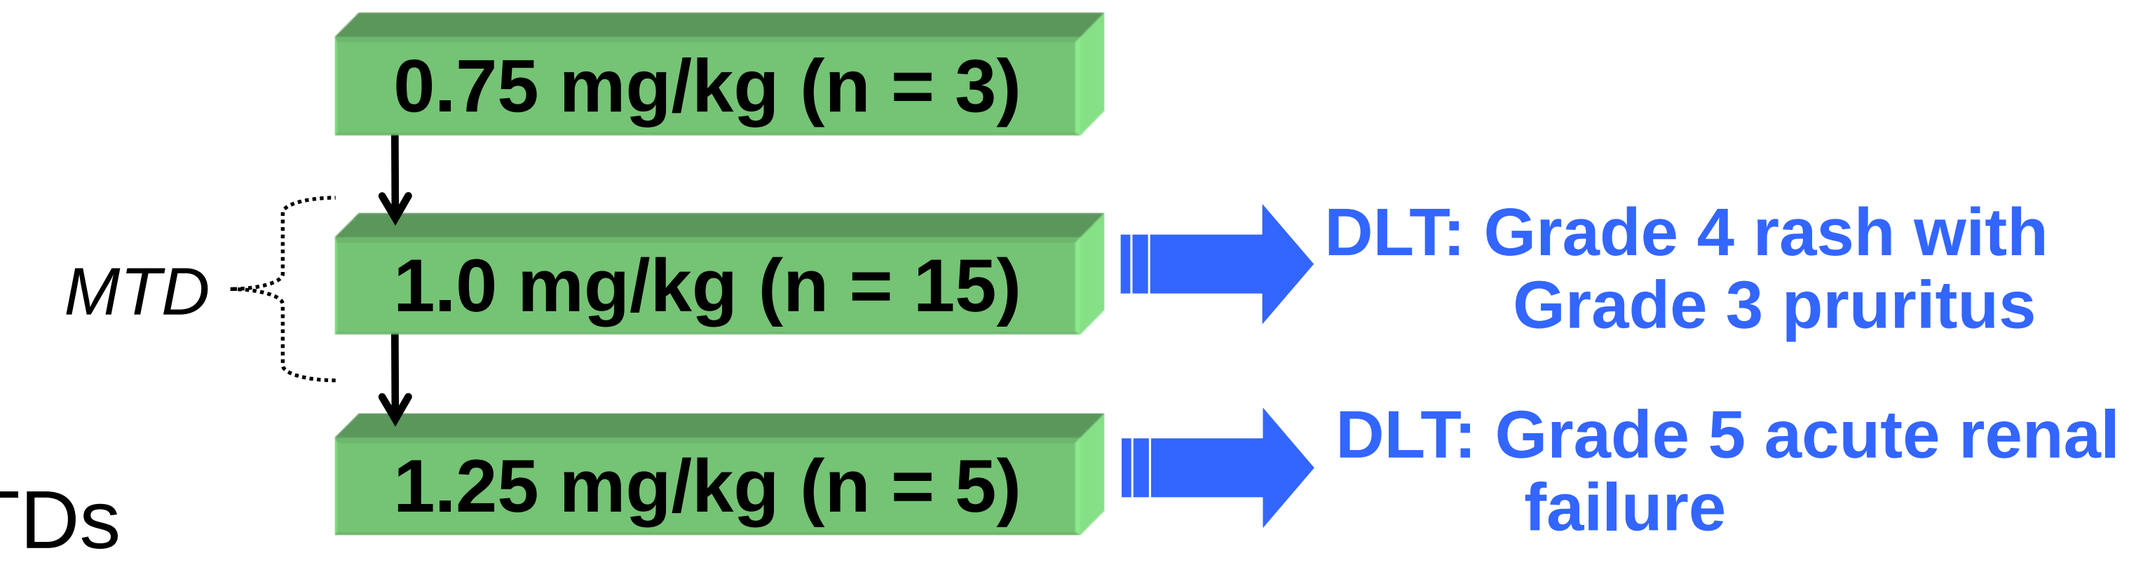
0.75 mg/kg (n = 3)
MTD
1.0 mg/kg (n = 15)
DLT: Grade 4 rash with Grade 3 pruritus
1.25 mg/kg (n = 5)
DLT: Grade 5 acute renal failure

ACTIVITY

Primary Activity Endpoint: Objective Response Rate (ORR) for the Phase II Expansion Cohort (q3w Schedule)

Statistical Plan: Study considered positive if ≥ 4 responses in 32 patients

	ORR
Primary Endpoint: Phase II population (1.88 mg/kg) [a]	5/34 (15%)

[a] Phase II population includes the 36 patients enrolled in the Phase II study portion. Two patients are excluded due to withdrawal of consent prior to post-treatment tumor assessment.

Maximum Tumor Shrinkage

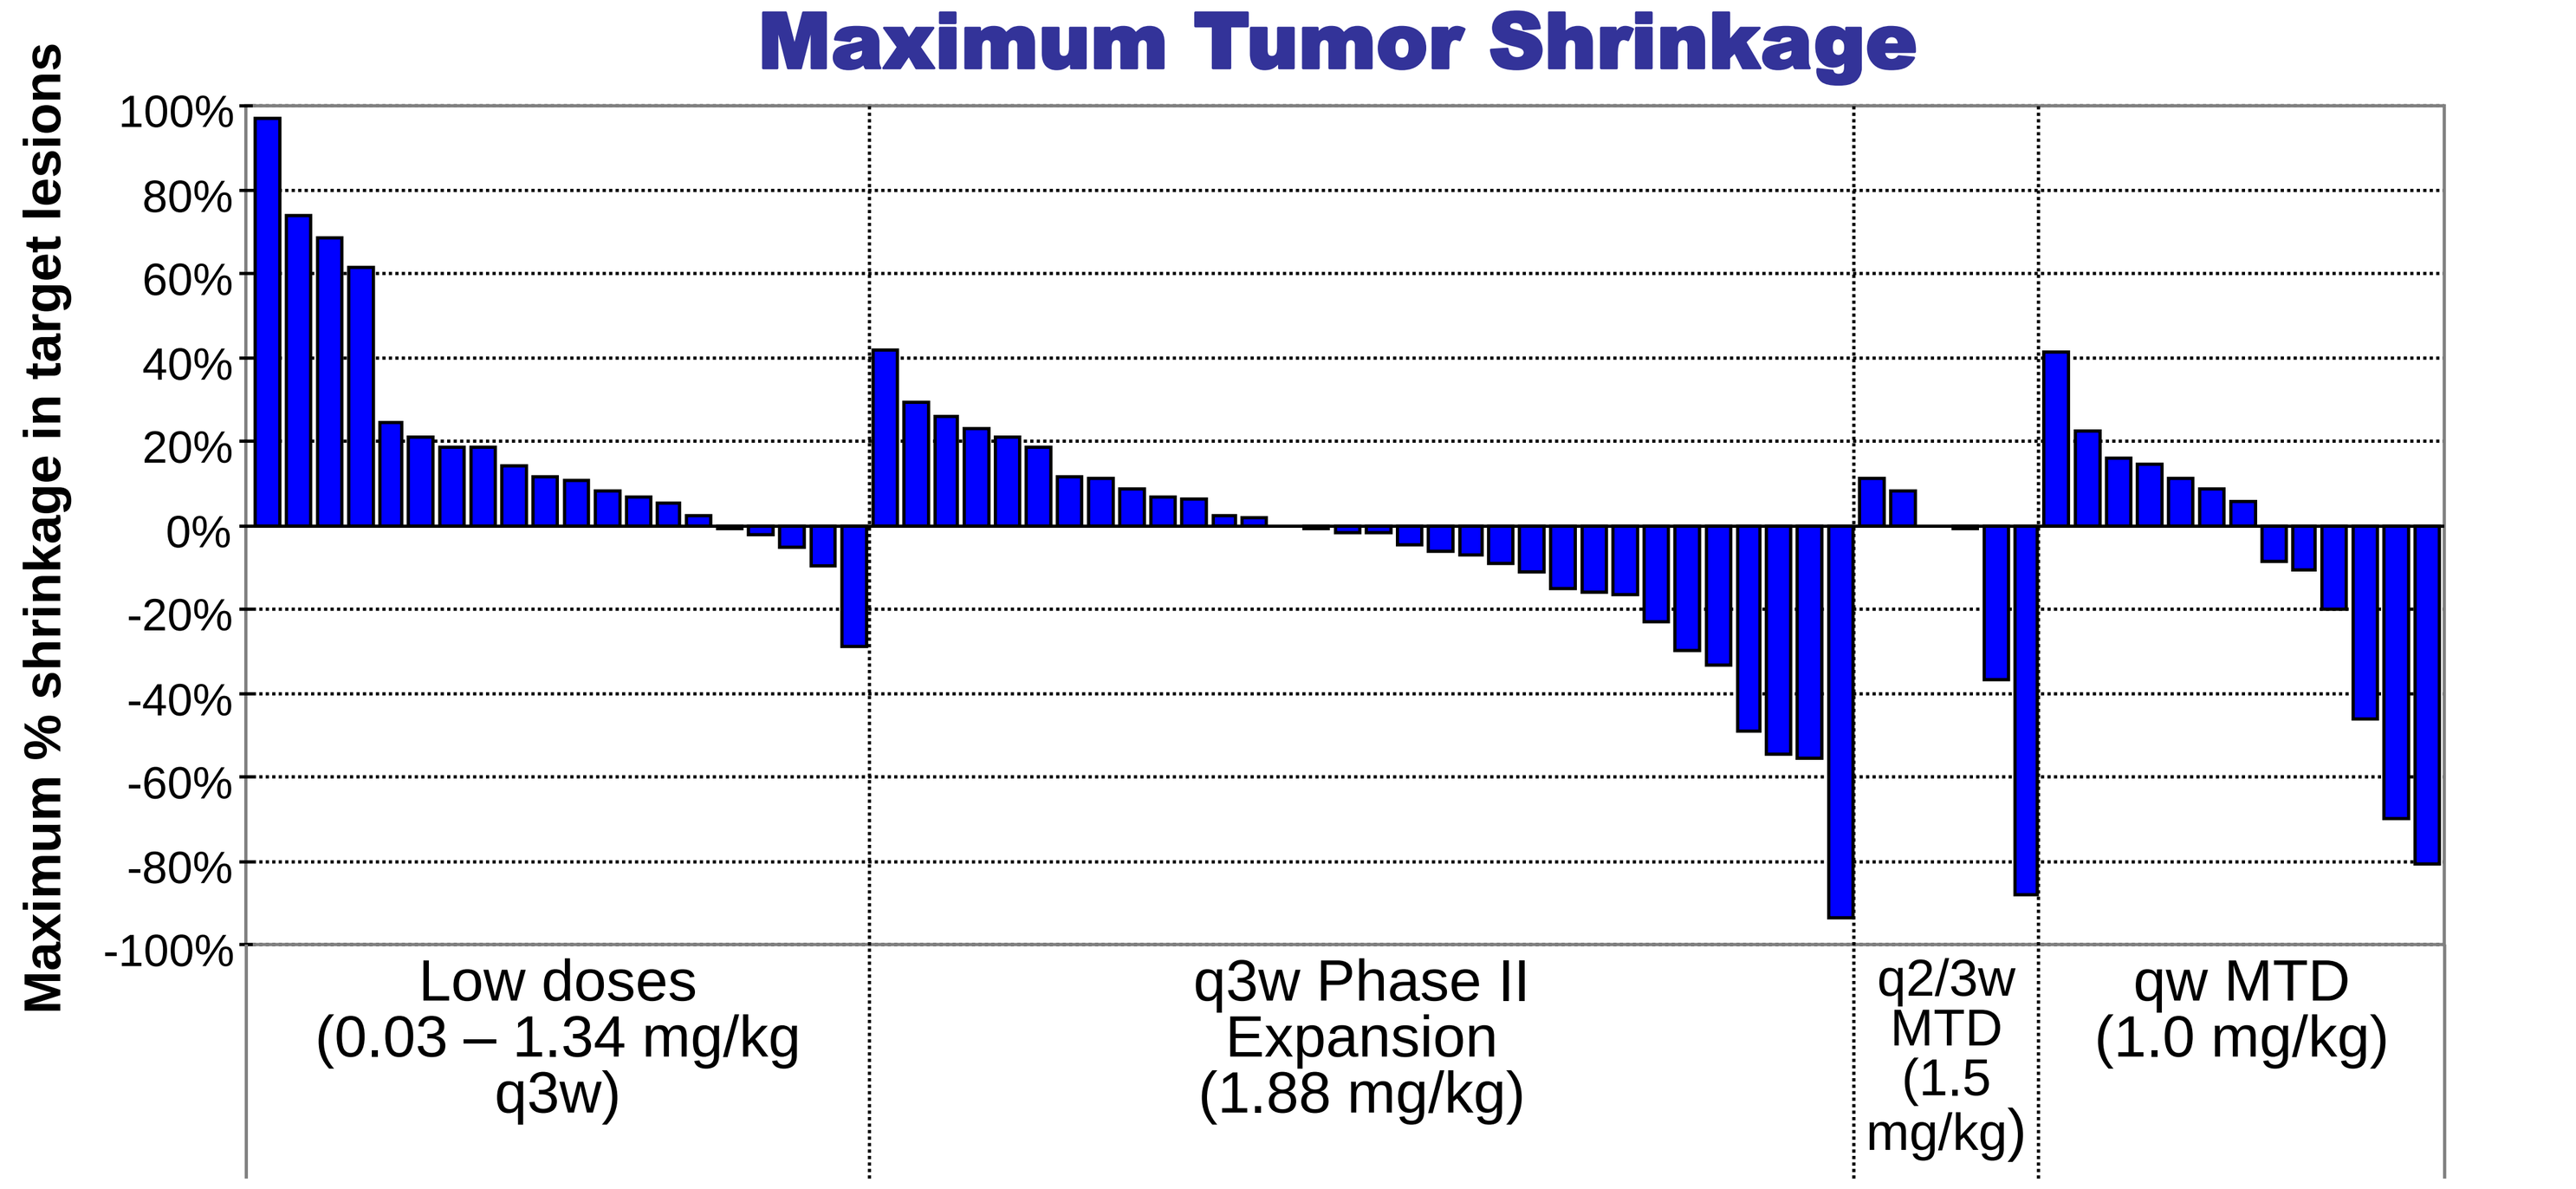
Maximum % shrinkage in target lesions
100%
80%
60%
40%
20%
0%
-20%
-40%
-60%
-80%
-100%
Low doses (0.03 – 1.34 mg/kg q3w)
q3w Phase II Expansion (1.88 mg/kg)
q2/3w MTD (1.5 mg/kg)
qw MTD (1.0 mg/kg)

Skin Rash Correlates with Outcome

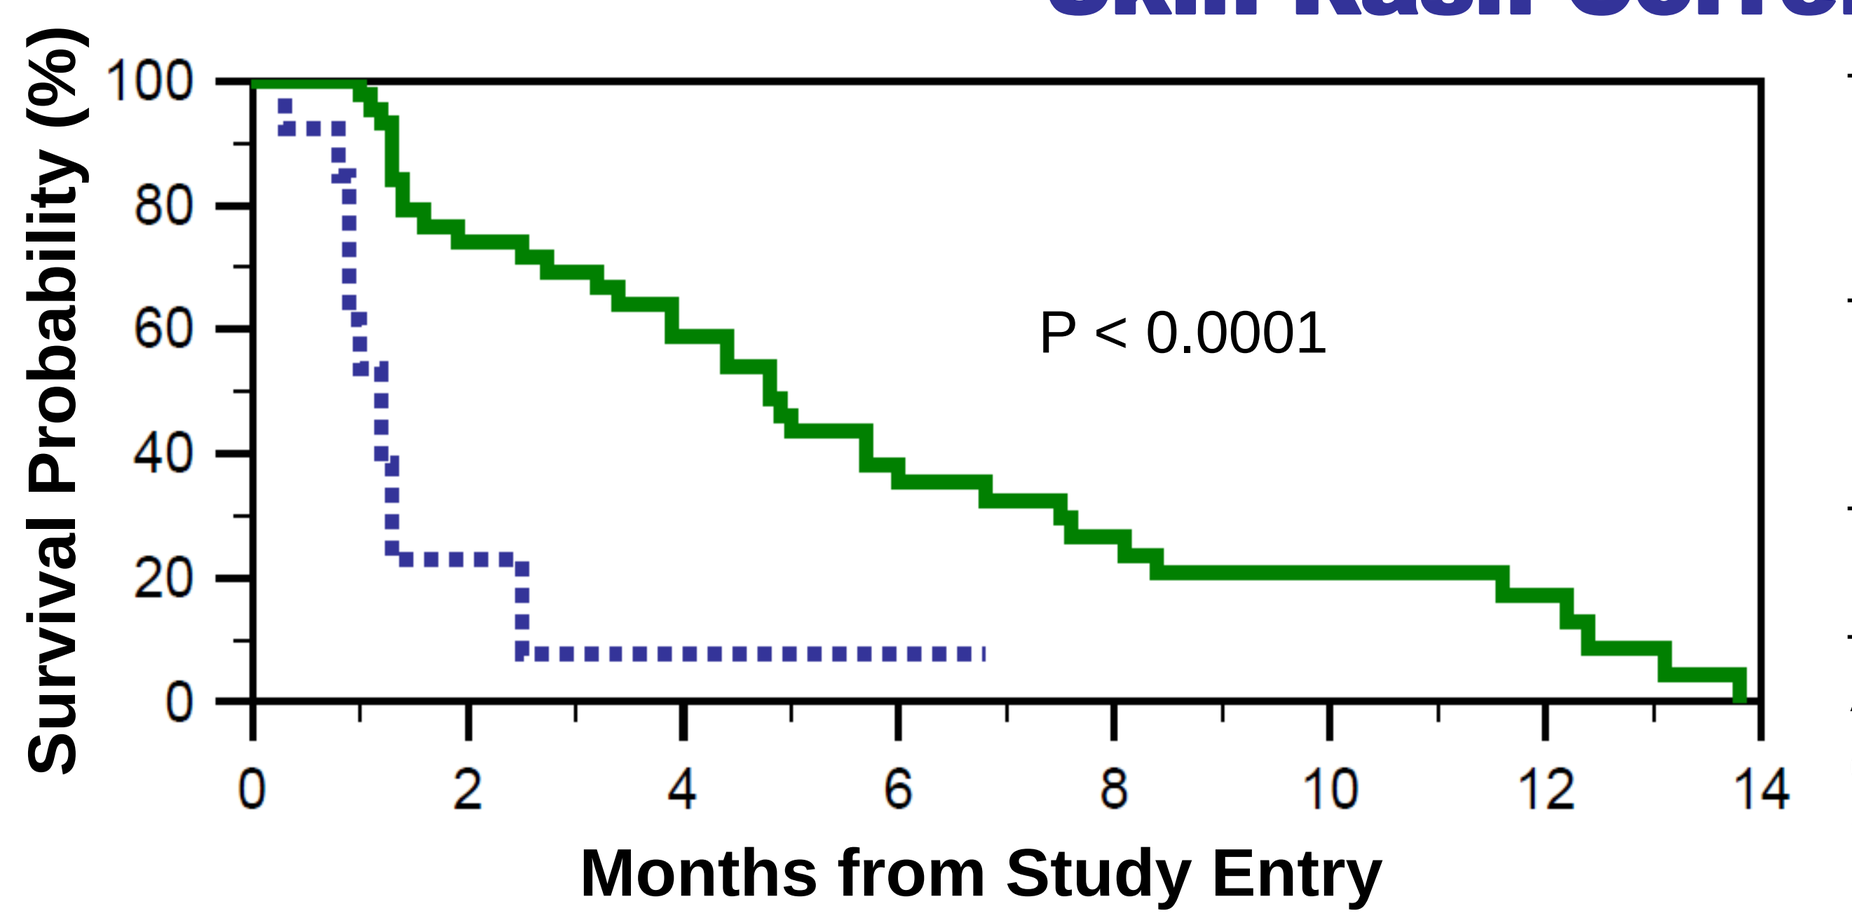
Survival Probability (%)
P < 0.0001
Months from Study Entry

	Median PFS (months)	ORR
Rash (n=44)	4.8	10/44 (23%)
No rash (n=13)	1.2	0%

Analyses include pooled patients from the q3w Phase II MTD expansion, q2/3w MTD and qw MTD. For 41/44 (93%) of patients who experienced rash, onset was within the first cycle.

Examples of Responses in GPNMB-Expressing Patients

Pt 4191: Strong GPNMB expression

- Treated at 1.5 mg/kg q2/3w
- Partial Response (88% shrinkage)
- PFS = 11.7 months (discontinued treatment due to neuropathy)

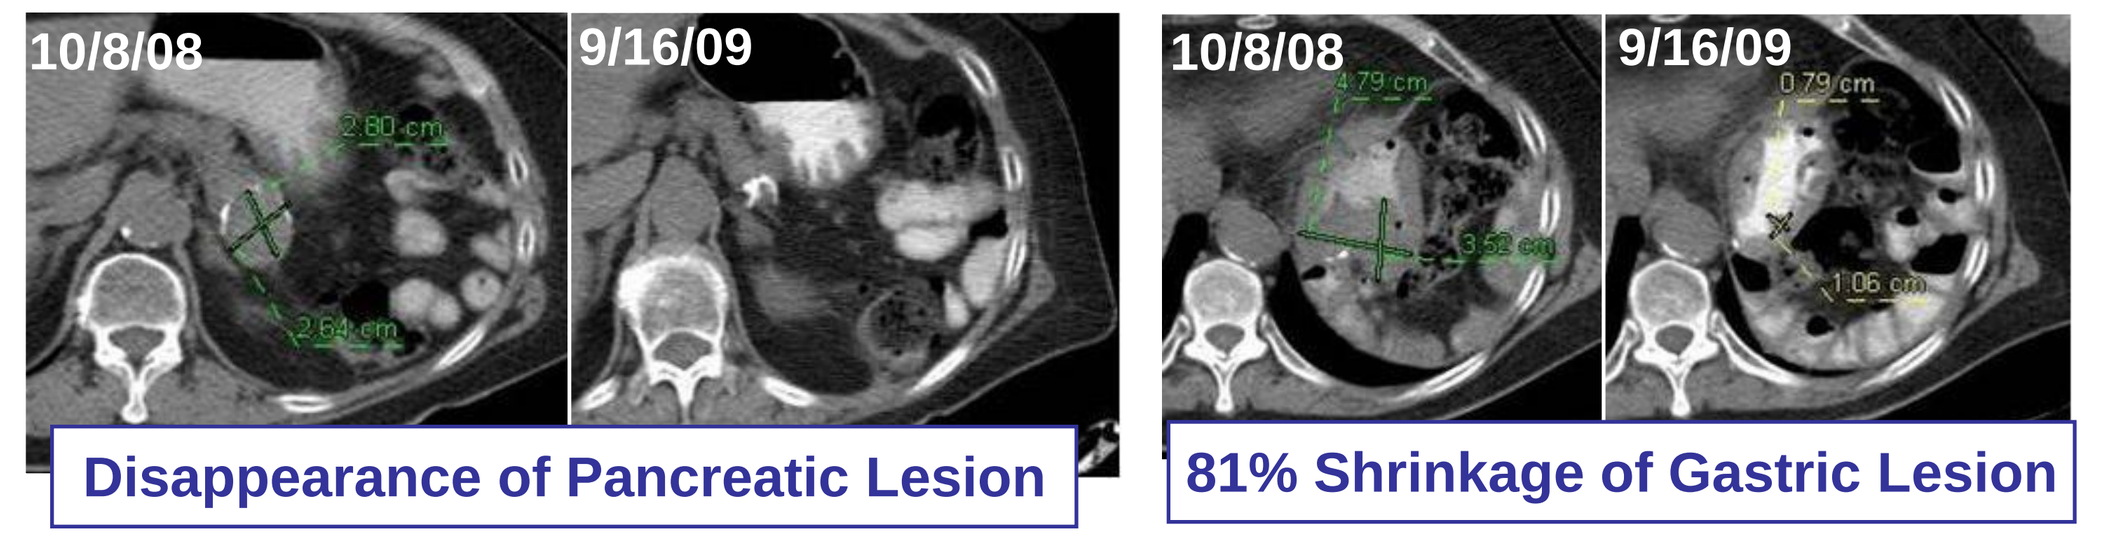
10/8/08
9/16/09
Disappearance of Pancreatic Lesion
10/8/08
9/16/09
81% Shrinkage of Gastric Lesion

Pt 2219: Strong GPNMB expression

- Treated at 1.0 mg/kg qw
- Partial Response (81% shrinkage), with disappearance of multiple thigh lesions
- PFS = 4.9 months

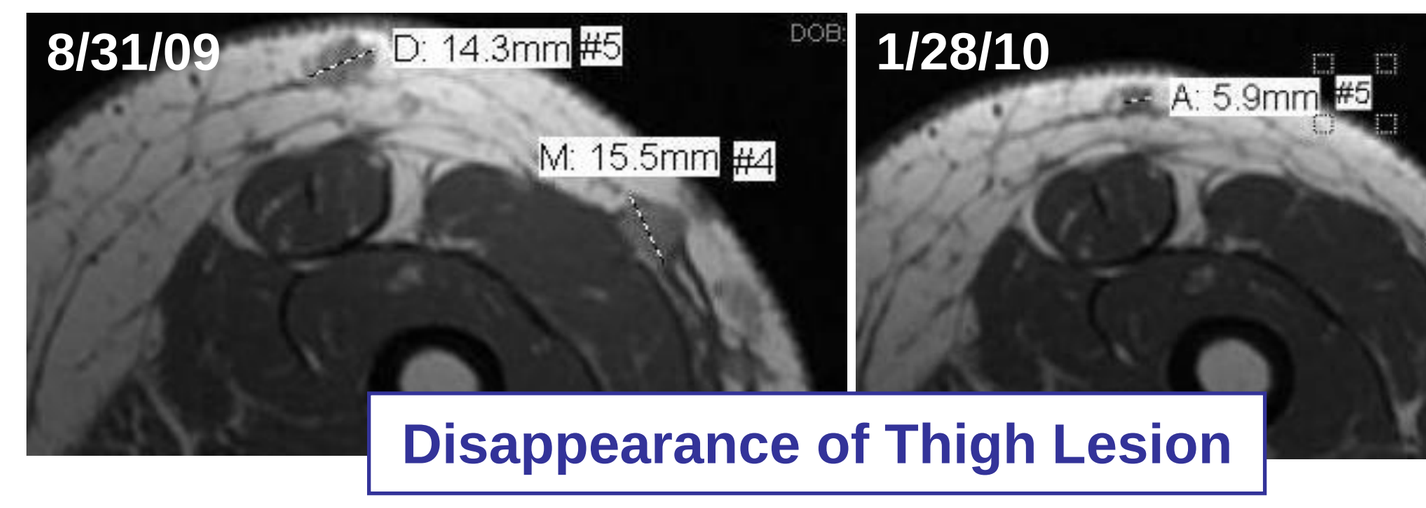
8/31/09
D: 14.3mm #5
M: 15.5mm #4
1/28/10
A: 5.9mm #5
#4
Disappearance of Thigh Lesion

Activity Parameters by Dosing Schedule and GPNMB Expression

	q3w Phase II MTD expansion (1.88 mg/kg)	q2/3w MTD (1.5 mg/kg)	qw MTD (1.0 mg/kg)	Strong (3+, 90%) GPNMB expression [a]
M1c Disease	18/34 (53%)	4/6 (67%)	12/15 (80%)	5/7 (71%)
Any tumor shrinkage	18/32 (56%) [b]	3/6 (50%)	6/15 (40%)	6/7 (86%)
Overall response rate (ORR)	5/34 (15%) [c]	2/6 (33%)	3/15 (20%)	2/7 (29%) [d]
PR or SD ≥ 12 weeks	20/34 (59%)	3/6 (50%)	5/15 (33%)	6/7 (86%)
Median PFS (months)	3.9	3.0	1.4 [e]	4.9

[a] Includes pooled patients from the q3w Phase II MTD expansion, q2/3w MTD and qw MTD.
[b] Excludes two additional patients without post-baseline assessment of measurable lesions.
[c] Including 1 unconfirmed PR (93% tumor shrinkage).
[d] ORR for patients with strong GPNMB expression and M1c disease = 2/5 (40%).
[e] One patient continues on treatment.

PATIENT CHARACTERISTICS, TOLERABILITY AND TREATMENT EXPOSURE

Demographic Characteristics (n=117)	
Male *(n [%])*	75 (64%)
Age *(years; median [range])*	62 (36-82)
Karnofsky PFS ≥ 90% *(n [%])*	102 (87%)
Disease Characteristics (n=117)	
Stage *(n [%])* III IV M1c	 13 (11%) 104 (89%) 59 (50%)
Baseline LDH > ULN *(n [%])*	49 (42%)
Duration of metastatic disease *(years; median [range])*	1.4 (0.05-10)
Prior anticancer regimens for metastatic disease *(median [range])*	1 (0-8)
Prior Therapies *(n [%])* Chemotherapy Immunotherapy Biochemotherapy CTLA-4 inhibitors Other investigational agents Kinase Inhibitors Vaccines	 61 (52%) 44 (38%) 12 (10%) 25 (21%) 26 (22%) 16 (14%) 12 (10%)

Toxicity	Occurring at any severity				≥ Grade 3 Severity (CTCAE)			
	q3w MTD (n=43)	q2/3w MTD (n=6)	qw MTD (n=15)	All (n=117)	q3w MTD (n=43)	q2/3w MTD (n=6)	qw MTD (n=15)	All (n=117)
Rash	70%	50%	60%	57%	26%	-	33%	20%
Fatigue	65%	67%	53%	56%	7%	17%	13%	8%
Pruritus	63%	33%	47%	49%	-	-	7%	3%
Alopecia	63%	33%	33%	40%	-	-	-	-
Diarrhea	47%	67%	20%	35%	2%	-	-	3%
Nausea	37%	50%	7%	29%	-	-	-	-
Anorexia	42%	33%	13%	26%	-	-	-	-
Neuropathy	35%	33%	60%	34%	5%	17%	27%	7%
Dysgeusia	33%	33%	13%	21%	-	-	-	-
Constipation	28%	33%	20%	19%	-	-	-	-
Neutropenia	26%	33%	13%	21%	14%	17%	13%	15%
Myalgia	12%	-	27%	13%	-	-	13%	3%
Arthralgia	7%	17%	7%	9%	-	-	7%	3%

- Table presents treatment-related adverse events occurring at any severity in >20%, or ≥ Grade 3 severity in >2% of patients. (Data cut-off as of May 7, 2010)
- Treatment-related deaths: Pneumococcal Sepsis (2.25 mg/kg q3w), Acute Renal Failure (1.25 mg/kg qw), and Toxic Epidermal Necrolysis (1.75 mg/kg q2/3w).

Treatment Parameter	q3w MTD (n=43)	q2/3w MTD (n=6)	qw MTD (n=15)
Duration of treatment (weeks) *(median [range])*	11 (0.1-58)	4 (1-49)	7 (0.1-38)
Cumulative dosing (mg/kg) *(median [range])*	7.5 (1.8-33)	6.1 (3.0-37)	6.0 (1.0-32)
Discontinued treatment for toxicity *(n [%])*	1 (2%)	2 (33%)	4 (27%)

PHARMACOKINETICS

- For the q3w schedule, half-life of antibody-drug conjugate (ADC) and total antibody (TA) increased with dose (ranges: 16-38 hours and 15-43 hours, respectively). Maximum MMAE and Tmax were dose proportional (ranges: 0.1-7.1 ng/mL and 20-169 hours, respectively).
- At doses of 1.88 mg/kg and higher in the q3w schedule, we observed correlations between severe rash and dose normalized exposure in cycle 1 to ADC (p=0.010) and TA (p=0.0129) but not MMAE (p=0.180).
- Across all dose levels and schedules, dose normalized exposure to TA in cycle 1 (AUCinf/dose) differed significantly for best response of PD, PR and SD (Kruskal-Wallis; p=0.0167) and correlated directly with objective response (Dunn's post test of PD vs. PR; p<0.05).

CONCLUSIONS

- **CDX-011, dosed either q3w, q2/3w or qw, is active in advanced melanoma.**
 - **The primary activity endpoint has been met.**
 - **Overall Response Rate (ORR) of ≥15% is promising.**
 - **There is evidence of increased activity with the q2/3w and qw schedules over the q3w schedule but this is accompanied by increased toxicity.**
- **Strong GPNMB expression and the development of rash, which may be related to the presence of GPNMB in the skin, are associated with greater progression-free survival.**
- **Further evaluation of CDX-011 in melanoma will include patients with strong GPNMB expression and combination therapy intended to upregulate GPNMB.**